Prevalence of Hepatitis B and D Virus Among a Nationally Representative Insured Population in the United States

Norah Terrault[1], Sacha Satram[2], Michael A Chattergoon[2], Christopher Dieyi[3], Jason Maynard[3], Radhika Trivedi[3], Richard Coppola[3], Nancy Reau[4]

[1]Keck School of Medicine of USC, Los Angeles, CA, USA; [2]Vir Biotechnology, San Francisco, CA, USA; [3]STATinMED LLC, Dallas, TX, USA; [4]Rush University Medical Center, Chicago, IL, USA

Introduction

- Hepatitis delta virus (HDV) is an incomplete human RNA virus that requires hepatitis B virus (HBV) surface antigen for replication.
- HDV is the most severe form of viral hepatitis[1] and presents as either a coinfection (simultaneously with HBV) or superinfection (already infected with HBV).[1]
- The true prevalence of HBV and HDV in the United States is unclear due to challenges in detection and testing, and many prevalence estimates rely on small studies from unique high-risk subpopulations.

Objective

- The objective of this study is to provide updated prevalence estimates of HBV and HDV using a large, US population-based administrative claims database.

Methods

- This cross-sectional study was conducted using medical claims from STATinMED RWD (Real World Data) Insights, an all-payer claims database which contains data for approximately 80% of the US-insured population.
- Patients were identified from January 1, 2014, to December 31, 2022. The entire database, consisting of approximately 278 million patients, was used as the base population.
- Disease cases with HBV and HDV were identified using 2 International Classification of Diseases, 9/10th Edition, Clinical Modification (ICD-9/10-CM) diagnosis codes at least 1 month apart to exclude rule-out diagnoses (i.e., patients screened but not diagnosed).
 - HBV codes: 070.20, 070.22, 070.30, 070.32, B16.2, B16.9, B18.1, B19.1, Z22.51
 - HDV codes: 070.21, 070.23, 070.31, 070.33, 070.42, 070.52, B16.0, B16.1, B18.0, B17.0
- Prevalence of HBV was defined for all patients in the entire database, and prevalence of HDV was defined among those with HBV.
 - Prevalence was also stratified by age: Adult (18 years) and pediatric (<18 years) patients.
- Demographics and socioeconomic characteristics (age, sex, race, ethnicity, geographic region, insurance type, income, and education) as of diagnosis date were measured.

Results

Prevalence

- A total of 548,722 patients met the case definition for HBV, which corresponded to a prevalence of 0.2%. **(Figure 1)**
 - The majority of patients with HBV were adults (99%) and 1% were pediatric patients.
- Among those with HBV, a total of 15,065 patients met the case definition for HDV, which corresponds to a prevalence of 2.8%.
- Patients with HDV had a slightly higher proportion of pediatric (3.3%) vs HBV, and the remaining 96.7% of HDV patients were adults.

Figure 1. Prevalence of HBV and HDV in the United States, 2014-2022

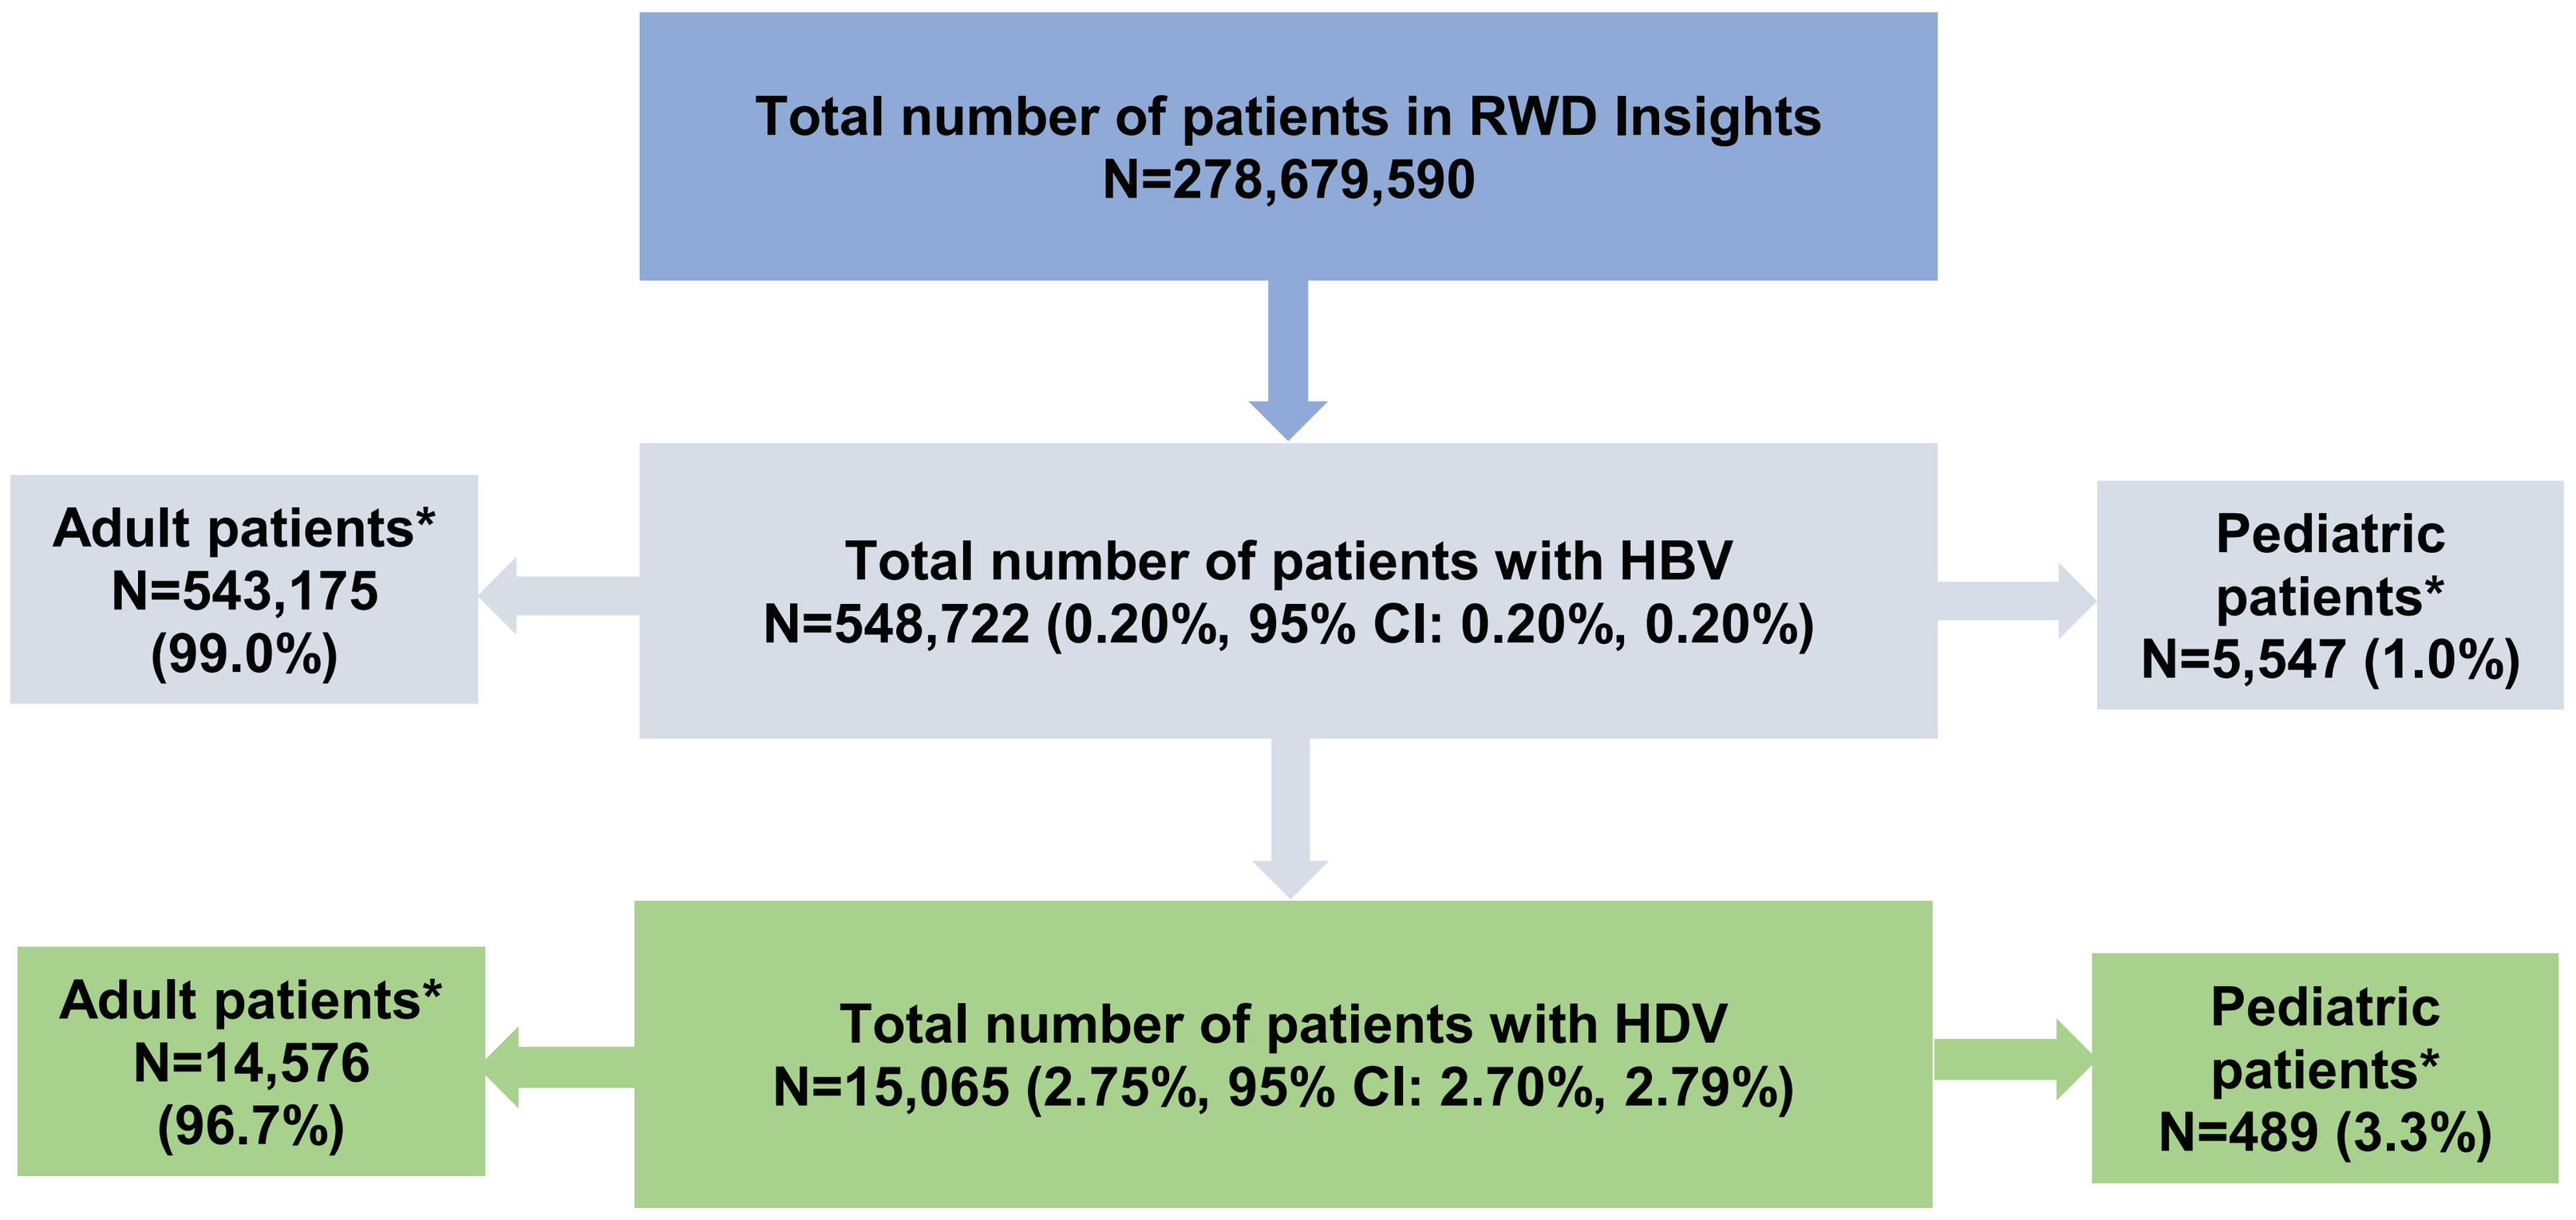
Total number of patients in RWD Insights
N=278,679,590
Adult patients*
N=543,175
(99.0%)
Total number of patients with HBV
N=548,722 (0.20%, 95% CI: 0.20%, 0.20%)
Pediatric patients*
N=5,547 (1.0%)
Adult patients*
N=14,576
(96.7%)
Total number of patients with HDV
N=15,065 (2.75%, 95% CI: 2.70%, 2.79%)
Pediatric patients*
N=489 (3.3%)

* Adult patients were aged 18+ years and pediatric patients were aged <18 years on their diagnosis date.
CI: confidence interval; HBV: hepatitis B virus; HDV: hepatitis D virus

Results

Figure 2. Prevalence of HDV in the United States by State, 2014-2022

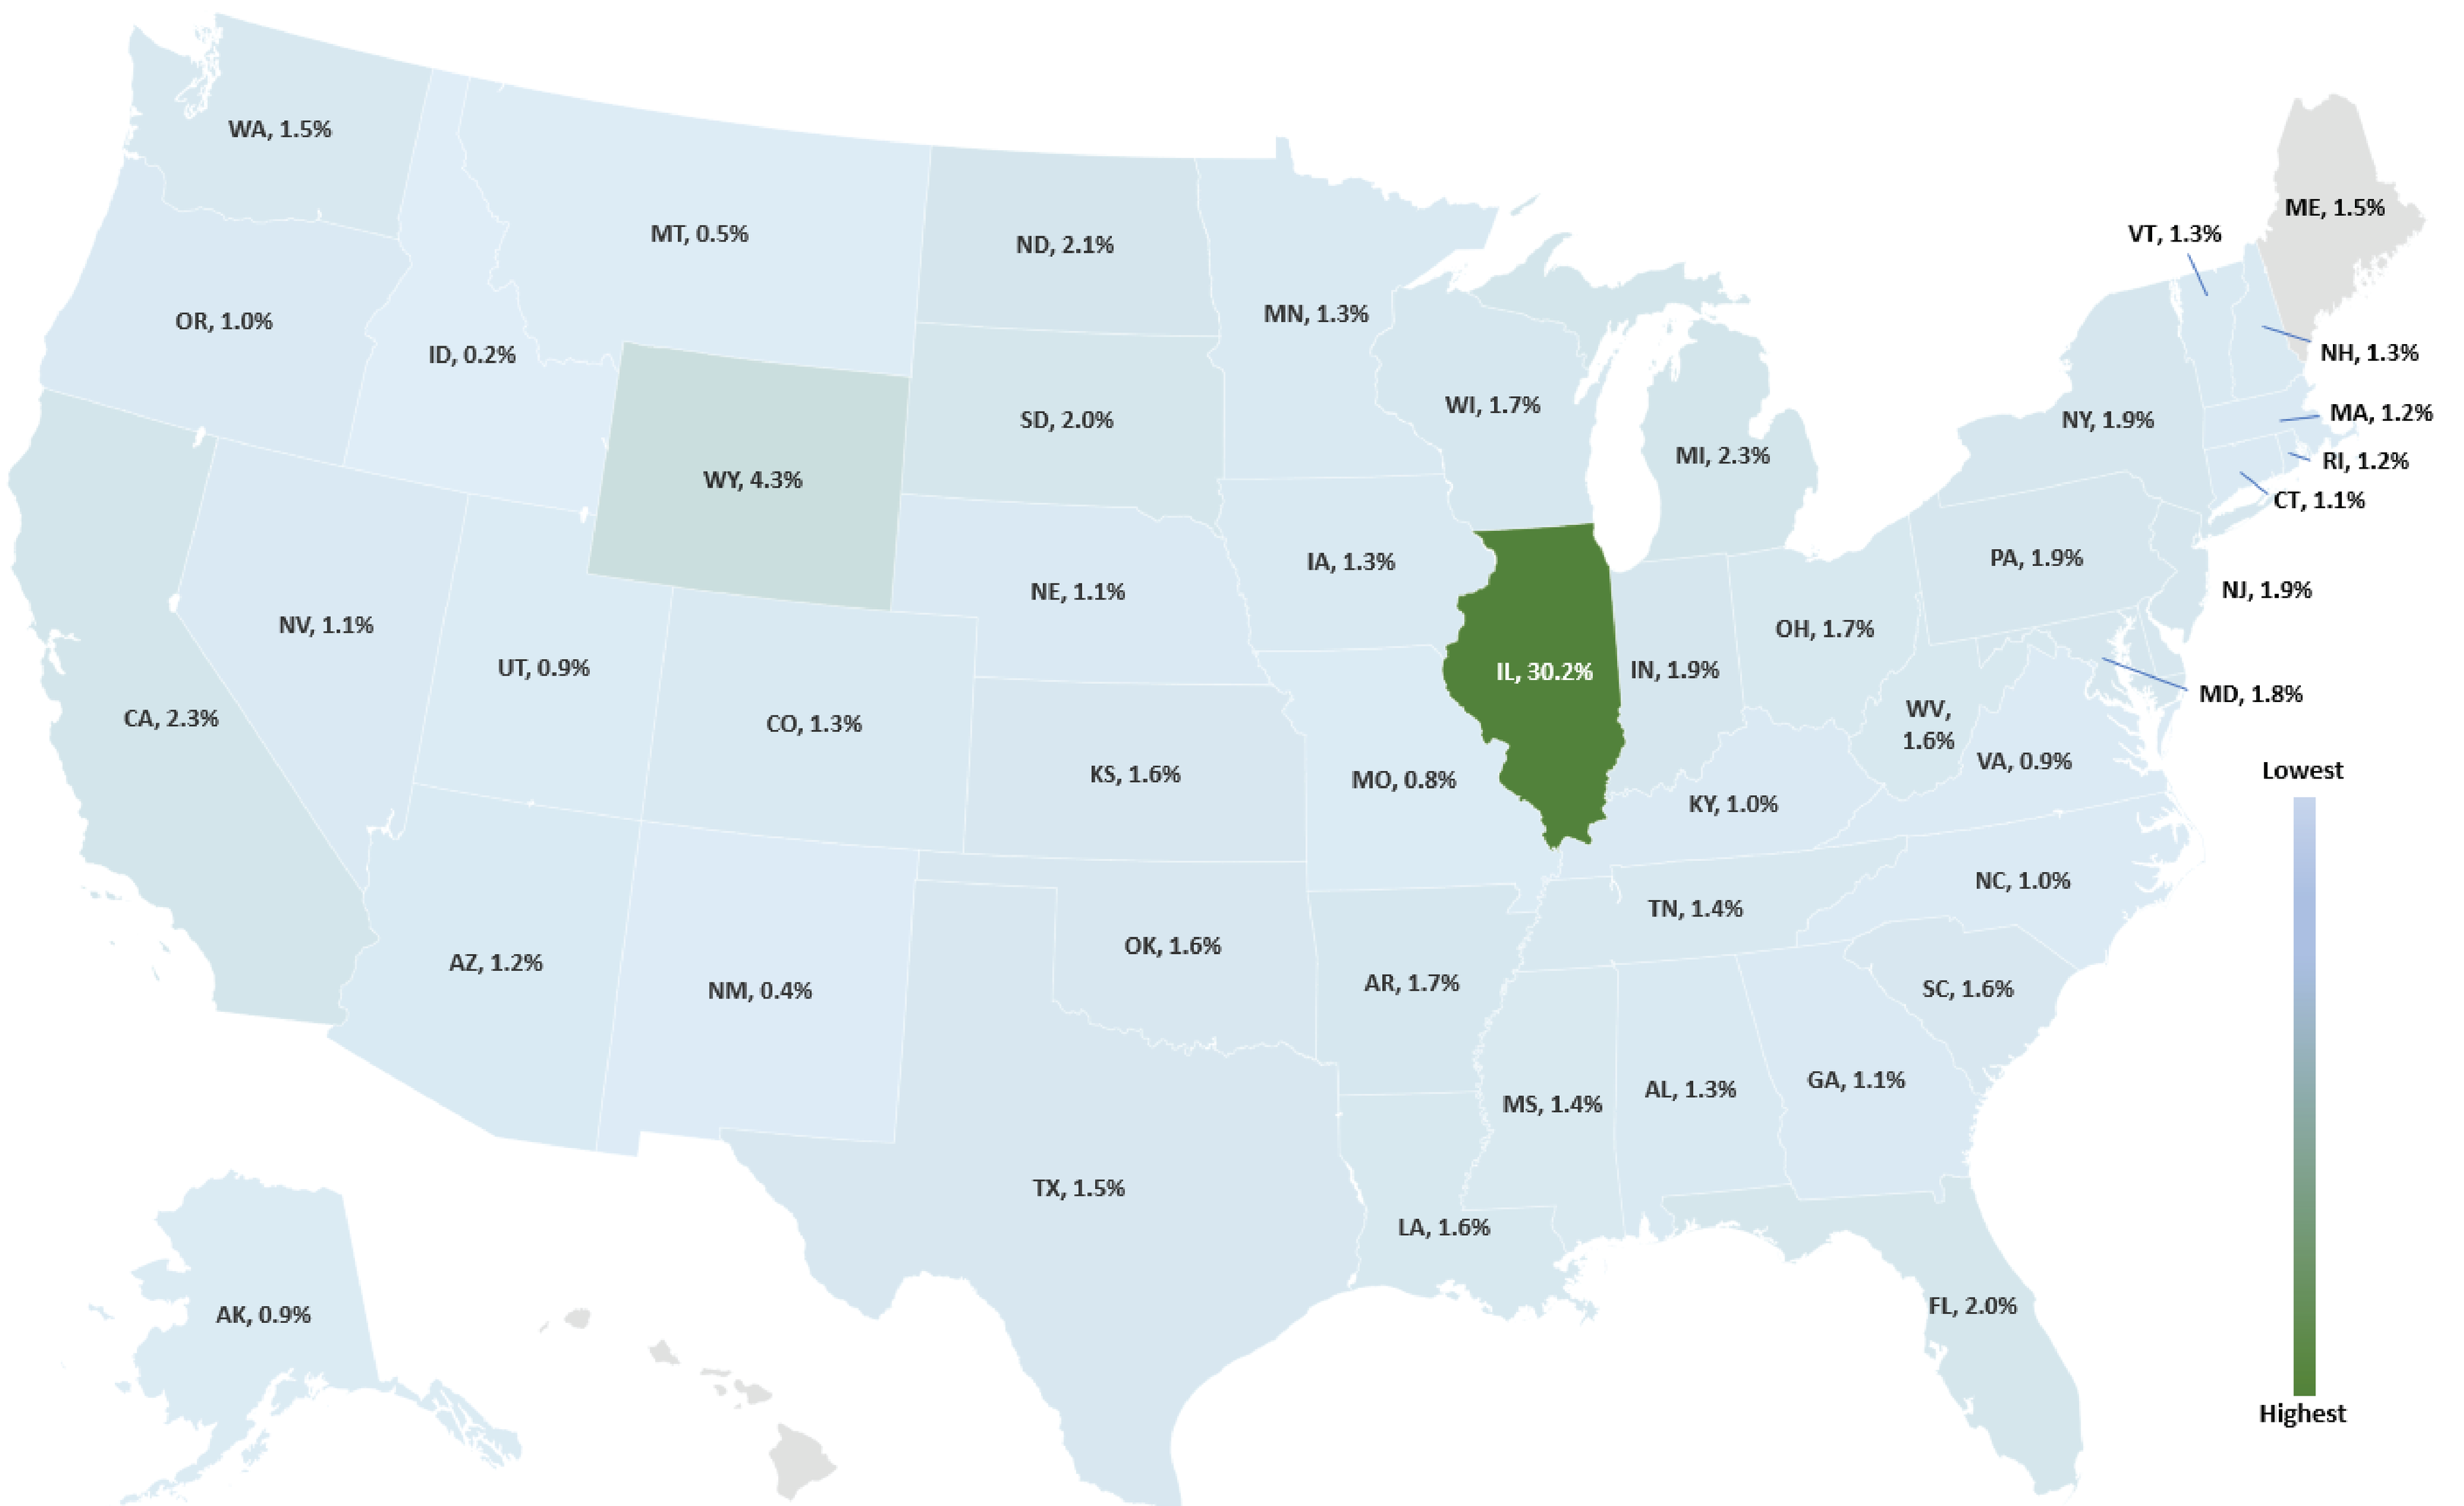
WA, 1.5%
OR, 1.0%
CA, 2.3%
ID, 0.2%
NV, 1.1%
MT, 0.5%
WY, 4.3%
UT, 0.9%
AZ, 1.2%
CO, 1.3%
NM, 0.4%
ND, 2.1%
SD, 2.0%
NE, 1.1%
KS, 1.6%
OK, 1.6%
TX, 1.5%
MN, 1.3%
IA, 1.3%
MO, 0.8%
AR, 1.7%
LA, 1.6%
WI, 1.7%
IL, 30.2%
MI, 2.3%
IN, 1.9%
OH, 1.7%
KY, 1.0%
TN, 1.4%
MS, 1.4%
AL, 1.3%
GA, 1.1%
FL, 2.0%
SC, 1.6%
NC, 1.0%
VA, 0.9%
WV, 1.6%
PA, 1.9%
NY, 1.9%
VT, 1.3%
ME, 1.5%
NH, 1.3%
MA, 1.2%
RI, 1.2%
CT, 1.1%
NJ, 1.9%
MD, 1.8%
AK, 0.9%
Lowest
Highest

The prevalence in Guam, Puerto Rico, and the Virgin Islands are not included in this map.

- HDV prevalence among patients with HBV, was highest in Illinois (30.2%), Wyoming (4.3%), California (2.3%), and Michigan (2.3%). **(Figure 2)**

Patient Characteristics

Table 1. Demographic Characteristics of Patients Diagnosed with HBV and HDV in the United States, 2014-2022

Demographic Characteristics	HBV N=548,722	HDV N=15,065	HDV Prevalence[1]
Age in years, mean (SD)	51.4 (15.0)	50.9 (16.5)	
Age groups, n (%)			
17 years and younger	5,547 (1.0%)	489 (3.2%)	8.8%
18 to 25 years	16,014 (2.9%)	704 (4.7%)	4.4%
26 to 35 years	70,707 (12.9%)	1,706 (11.3%)	2.4%
36 to 45 years	102,576 (18.7%)	2,378 (15.8%)	2.3%
46 to 54 years	109,426 (19.9%)	2,869 (19.0%)	2.6%
55 to 64 years	124,253 (22.6%)	3,534 (23.5%)	2.8%
65 to 74 years	86,666 (15.8%)	2,313 (15.4%)	2.7%
75 years and older	33,533 (6.1%)	1,072 (7.1%)	3.2%
Sex, n (%)			
Male	296,029 (53.9%)	7,794 (51.7%)	2.6%
Female	252,693 (46.1%)	7,271 (48.3%)	2.9%
Insurance type, n (%)			
Medicare	154,045 (28.1%)	4,245 (28.2%)	2.8%
Medicaid	232,356 (42.3%)	8,364 (55.4%)	3.6%
Commercial	151,923 (27.7%)	2,369 (15.7%)	1.6%
Government	10,398 (1.9%)	87 (0.6%)	0.8%

*Prevalence was estimated as patients with HDV divided by patients with HBV.
HBV: hepatitis B virus; HDV: hepatitis D virus; SD: standard deviation

- The average age of patients in the HBV (51.4 years) and HDV (50.9 years) cohorts was similar; the highest proportion of patients were 55 to 64 years of age (22.6% and 23.5%), respectively. **(Table 1)**
- While there were more men than women infected with both HBV and HDV; slightly more male patients had HBV compared to HDV (53.9% and 51.7%, respectively).
- In both cohorts, most patients were enrolled in a Medicaid health plan. **(Table 1)**

Results

Table 2. Socioeconomic Characteristics of Patients Diagnosed with HBV and HDV in the United States, 2014-2022

Socioeconomic Characteristics	HBV	HDV	HDV Prevalence*
Race, n (%)	188,648	5,786	
White	90,343 (47.9%)	2,453 (42.4%)	2.7%
Black	30,656 (16.3%)	1,926 (33.3%)	6.3%
Asian	66,456 (35.2%)	1,364 (23.6%)	2.1%
Other	1,193 (0.6%)	43 (0.7%)	3.6%
Ethnicity, n (%)	173,289	5,193	
Hispanic	26,254 (15.2%)	966 (18.6%)	3.7%
Not Hispanic	147,035 (84.8%)	4,227 (81.4%)	2.9%
Household income (USD $), n (%)	188,929	5,810	
25,000 and lower	72,138 (38.2%)	2,697 (46.4%)	3.7%
25,001 - 50,000	49,895 (26.4%)	1,477 (25.4%)	3.0%
50,001 - 75,000	28,256 (15.0%)	726 (12.5%)	2.6%
75,001 - 100,000	15,298 (8.1%)	385 (6.6%)	2.5%
101,000 and higher	23,342 (12.4%)	525 (9.0%)	2.3%
Education level, n (%)	120,020	3,640	
Some High School	2,236 (0.4%)	100 (2.7%)	4.5%
High School	65,130 (11.9%)	2,165 (59.5%)	3.3%
College	32,847 (6.0%)	891 (24.5%)	2.7%
Post Graduate	19,807 (3.6%)	484 (13.3%)	2.4%

*Prevalence was estimated as patients with HDV divided by patients with HBV. These proportions were estimated among patients with non-missing information.

- While most patients with non-missing race information were white **(Table 2)**, there was a greater proportion of black patients in the HDV group (33.3%) compared to HBV group (16.3%).
 - Black patients had the highest prevalence of HDV (6.3%) compared to all other race categories (2.1-3.6%).
- More Asian patients were in the HBV group compared to the HDV group (35% vs 24%).
- Most of the sample were of non-Hispanic ethnicity (81-85%).
- Where information on income and education were available, most patients were in the lowest household income category of $25,000 or lower and completed up to high school level of education. **(Table 2)**

Limitations

- Claims data are subject to misclassification bias in that the presence of a diagnosis code may not always represent actual diagnosis.
 - Bias was mitigated by using 2 diagnoses codes to rule out patients that were screened for HBV and HDV but were not diagnosed.
- The reported prevalence may only apply to the insured US population and may not be generalized to the uninsured US population.
- Due to inadequate screening procedures in real-world clinical practice, the true prevalence of HBV and HDV may be underestimated and thus our study results represent the "diagnosed" prevalence of disease in the United States.

Conclusions

- **This is the largest study to date on the prevalence of HBV and HDV in the United States and provides further support of a previously reported 3% overall prevalence of HDV among patients with HBV.[1,3]**
- **The US prevalence of HDV is highest in Illinois (30.2%) compared to all other states (<5%), higher among black patients compared to other racial groups, and among those <65 years of age.**
- **A robust definition of HBV and HDV that required confirmation of at least 2 diagnostic codes on medical claims is a strength of this study and adds to the existing sparse literature on the prevalence of HDV in the United States.**

References

1. Kushner T, Serper M, Kaplan DE. Delta hepatitis within the Veterans Affairs medical system in the United States: Prevalence, risk factors, and outcomes. *J Hepatol.* 2015;63(3):586–592.
2. Roberts H, Ly KN, Yin S, et al. Prevalence of HBV infection, vaccine-induced immunity, and susceptibility among at-risk populations: US households, 2013-2018. *Hepatol.* 2021;74(5):2353-65.
3. Terrault NA, Ghany MG. Enhanced screening for Hepatitis D in the USA: Overcoming the Delta blues. *Dig Dis Sci.* 2021;66(8):2483-5.

Disclosures

NT received grant/research support from Gilead Sciences. SS and MAC are employees of and own stock in Vir Biotechnology. CD, RC, and RT are employees of STATinMED, LLC, a paid consultant for Vir Biotechnology. JM was an employee of STATinMED, LLC, at the time of the study. NR is a paid consultant for Gilead Sciences.

Presented at the American Association for the Study of Liver Diseases (AASLD) The LIver Meeting, November 10-14, 2023